SGO ANNUAL MEETING ON WOMEN'S CANCER TAMPA, FL · 2023

PATIENTS • PURPOSE • PROGRESS

291

TRIAL IN PROGRESS: A PHASE 2 TRIAL OF NIROGACESTAT, A γ-SECRETASE INHIBITOR, IN PATIENTS WITH RECURRENT OVARIAN GRANULOSA CELL TUMORS (NCT05348356)

Panagiotis A. Konstantinopoulos, MD;[1] Jocelyn A. Lewis, DO;[2] Todd Shearer, PhD;[2] Heather O'Brien, BSc;[2] Caroline J. Breitbach, PhD;[2] Shinta Cheng, MD[2]

[1]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, USA; [2]SpringWorks Therapeutics, Inc., Stamford, CT, USA

INTRODUCTION

- Ovarian granulosa cell tumors (OvGCT) represent 5-7% of all ovarian cancers and account for 70% of ovarian sex cord-stromal tumors[1]
 - Surgery is the initial treatment with postoperative therapy considered for patients with more advanced disease
 - In relapsed patients, chemotherapeutic regimens have shown only modest activity[2-4]
- Notch signaling, the *FOXL2* C134W mutation, and dysregulation of the phosphatidylinositol 3-kinases/protein kinase B (PI3K/AKT) pathway are integral to the molecular pathogenesis of OvGCT[5] (**Figure 1**)
- The investigational agent nirogacestat (PF-03084014) is a potent, selective, reversible, noncompetitive inhibitor of γ-secretase, an enzyme that plays a pivotal role in Notch signaling[6-8]
 - Treatment with nirogacestat might be expected to impair granulosa cell proliferation in OvGCT by inhibiting proliferation signaling through the Notch intracellular domain (NICD), compensating for impaired induction of apoptosis by the *FOXL2* C134W mutation, and restoring negative regulation of PI3K and pAKT
- The primary objective of the current trial is to determine the anti-tumor activity of nirogacestat in adult patients with relapsed/refractory OvGCT

Figure 1. Schematic representation of the cell signaling pathways in OvGCT development.

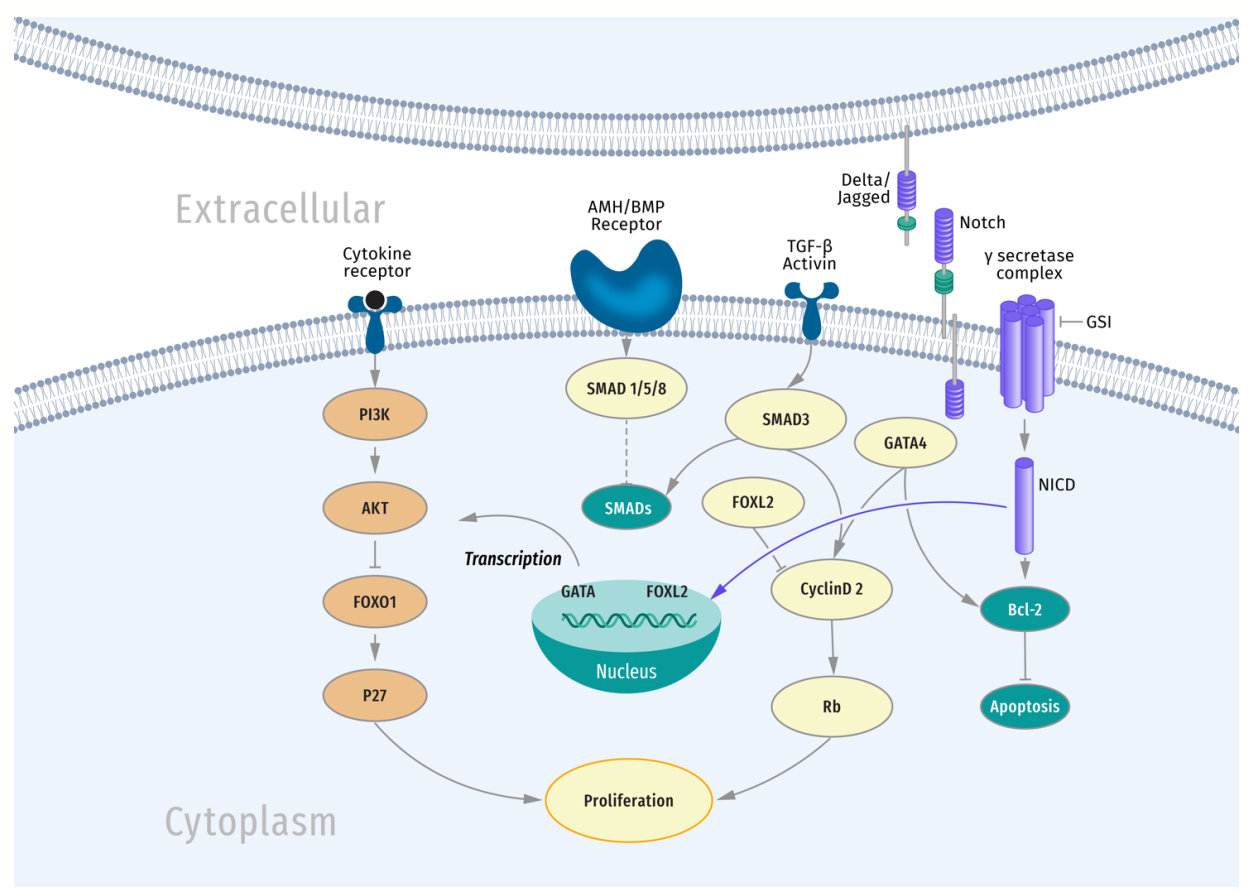
Extracellular
Cytokine receptor
AMH/BMP Receptor
TGF-β Activin
Delta/ Jagged
Notch
γ secretase complex
GSI
PI3K
AKT
FOXO1
P27
SMAD 1/5/8
SMADs
SMAD3
FOXL2
GATA4
NICD
Transcription
GATA
FOXL2
Nucleus
CyclinD 2
Rb
Bcl-2
Apoptosis
Proliferation
Cytoplasm

FOXL2 (forkhead box L2) is a transcription factor involved in the regulation of hormone production, cell cycle, and apoptosis. The somatic mutation *FOXL2* C134W occurs in > 97% of patients with adult OvGCT and may lead to dysregulation of multiple cellular processes. In Notch signaling, ligands Delta or Jagged bind to the Notch extracellular domain and activate cleavage by the γ-secretase complex to release the NICD (Notch intracellular domain). The NICD translocates to the nucleus to stimulate gene expression. Notch signaling also interacts with PI3K/AKT (phosphatidylinositol 3-kinases/protein kinase B) signaling. Activation of the AKT pathway via PI3K blocks apoptosis through inhibition of FOXO1 (forkhead box O1) and activation of Bcl-2 (B-cell lymphoma 2) transcription. GSI (γ-secretase inhibitors) can prevent release of the NICD. Adapted from Li et al.[5]

TRIAL OVERVIEW

Study Design

- Multi-center, single-arm, Phase 2, open-label
- Eligible patients are females ≥18 years of age with histologically confirmed recurrent adult-type OvGCT with measurable disease by Response Evaluation Criteria in Solid Tumors (RECIST) v1.1
 - Key inclusion and exclusion criteria are shown in **Table 1**
- Treatment is with nirogacestat 150 mg PO BID in continuous 28-day cycles until death, disease progression, or study discontinuation
 - All patients will be followed for ≥2 years
- Statistical analysis employs a Bayesian strategy allowing continuous monitoring to evaluate objective response rate (ORR) (**Figure 2**)

Figure 2. Study design with continuous monitoring for futility analysis.

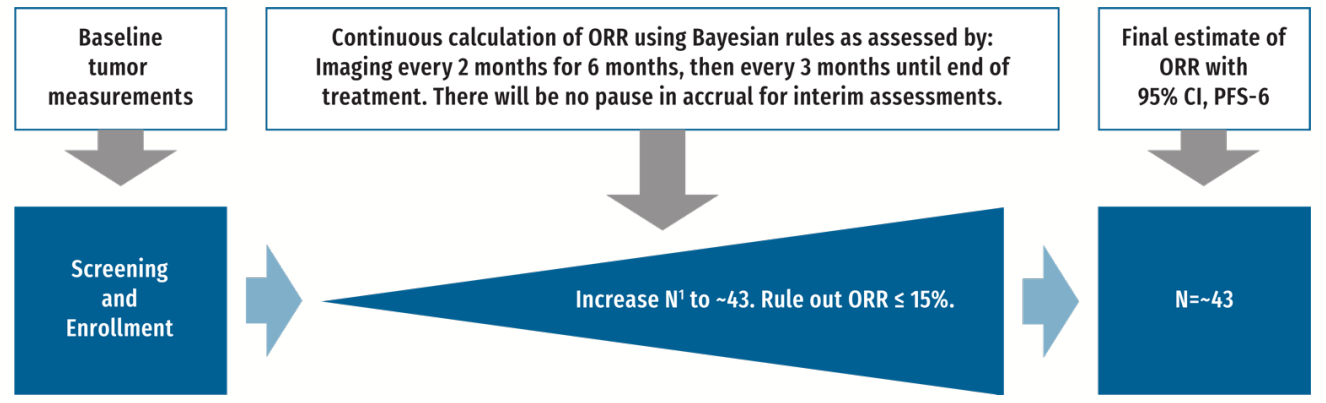
Baseline tumor measurements
Continuous calculation of ORR using Bayesian rules as assessed by: Imaging every 2 months for 6 months, then every 3 months until end of treatment. There will be no pause in accrual for interim assessments.
Final estimate of ORR with 95% CI, PFS-6
Screening and Enrollment
Increase N[1] to ~43. Rule out ORR ≤ 15%.
N=~43

ORR, objective response rate; PFS-6, 6-month progression-free survival.

- Approximately 43 patients are expected to enroll
- Currently recruiting at 10 sites in the US (**Figure 3**)

Figure 3. Currently recruiting study sites.

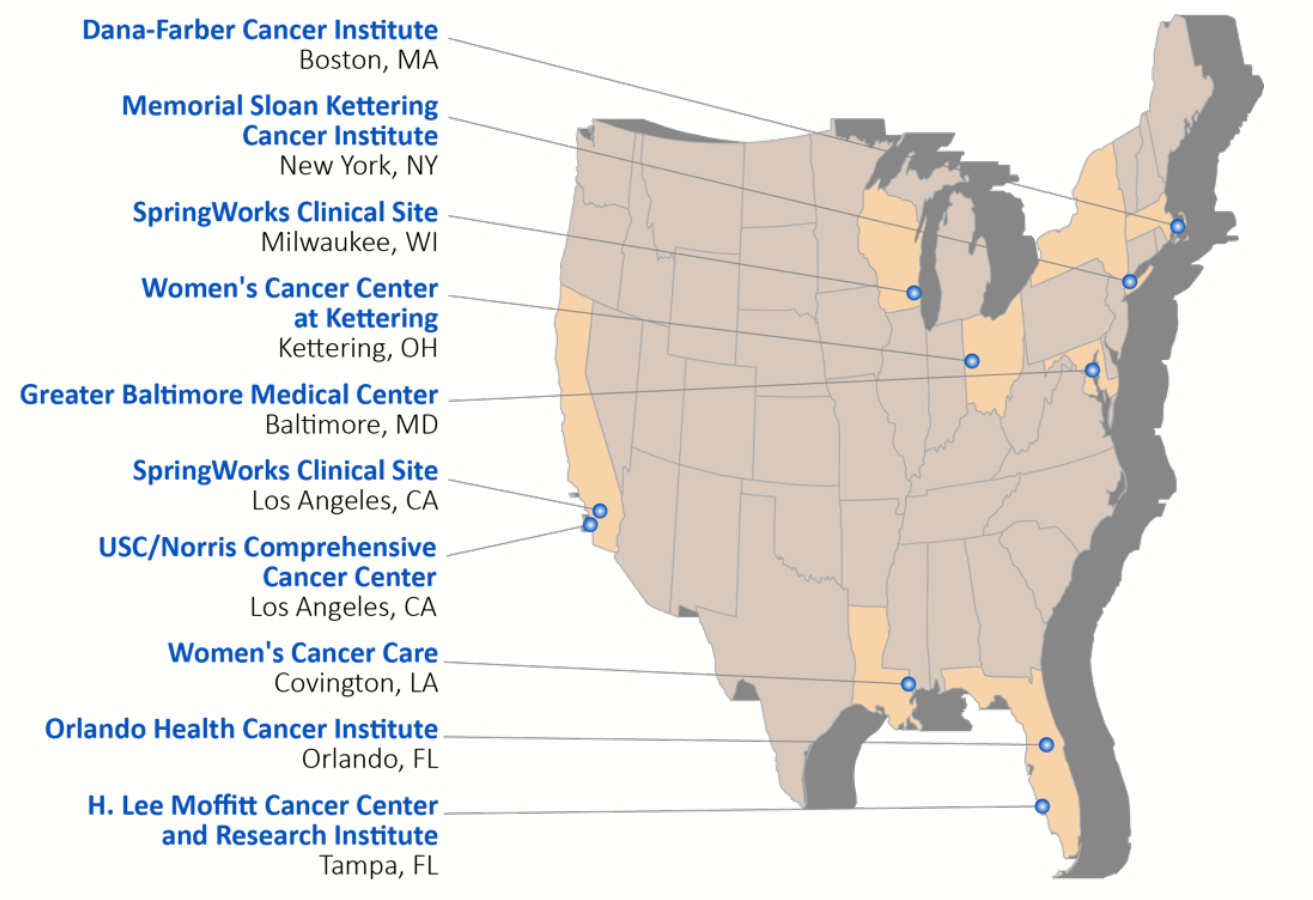
Dana-Farber Cancer Institute
Boston, MA
Memorial Sloan Kettering Cancer Institute
New York, NY
SpringWorks Clinical Site
Milwaukee, WI
Women's Cancer Center at Kettering
Kettering, OH
Greater Baltimore Medical Center
Baltimore, MD
SpringWorks Clinical Site
Los Angeles, CA
USC/Norris Comprehensive Cancer Center
Los Angeles, CA
Women's Cancer Care
Covington, LA
Orlando Health Cancer Institute
Orlando, FL
H. Lee Moffitt Cancer Center and Research Institute
Tampa, FL

Table 1. Key inclusion and exclusion criteria.

Inclusion Criteria	Exclusion Criteria
• Females ≥18 years of age	• Signs of bowel obstruction requiring parenteral nutrition, malabsorption syndrome, or preexisting gastrointestinal conditions that may impair absorption of nirogacestat
• Histologically confirmed recurrent adult-type OvGCT	• History of major cardiac or thromboembolic event within 6 months of signing informed consent
• Documented radiological evidence of relapse after ≥1 systemic therapy that is not amenable to surgery or radiation, and measurable disease by RECIST v1.1 criteria	• Abnormal QT interval at screening, or congenital or acquired long QT syndrome, or history of additional risk factors for Torsades de Pointes
• ECOG performance score of 0, 1, or 2 at screening	• Current or chronic history of liver disease or known hepatic or biliary abnormalities
• Adequate bone marrow, renal, and hepatic function	• Treatment for OvGCT within 28 days (or 5 half-lives, whichever is longer) with anti-angiogenic therapy, hormonal therapy, chemotherapy, immunotherapy, targeted therapy, or any investigational treatment

ECOG, Eastern Cooperative Oncology Group; OvGCT, ovarian granulosa cell tumor; RECIST, Response Evaluation Criteria in Solid Tumors.

Outcome Evaluations

- Study objectives and outcome evaluations are summarized in **Table 2**
- Efficacy:
 - The primary efficacy endpoint is ORR by RECIST v1.1
 - Secondary endpoints include 6-month progression-free survival and 2-year overall survival, change from baseline in the Functional Assessment of Cancer Therapy – Ovarian Symptom Index, and duration of response
- Safety: Incidence of treatment-emergent adverse events, changes in clinical laboratory parameters, vital signs, physical examination findings, and electrocardiograms
- Pharmacokinetic: Serum concentrations to evaluate systemic exposure of nirogacestat
- Exploratory: Changes in relevant biomarkers and tumor genomic alterations

Table 2. Study objectives and outcome evaluations.

Objectives	Endpoints
Primary Objective	**Primary Endpoint**
• To determine the anti-tumor activity of nirogacestat in adult participants with relapsed/ refractory OvGCT	• ORR, defined as the proportion of participants with CR + PR by RECIST v1.1 criteria
Secondary Objectives	**Secondary Endpoints**
• To determine if nirogacestat delays disease progression or death in OvGCT	• Estimate of proportion of participants with PFS-6 by RECIST v1.1 criteria
• To describe overall survival in participants treated with nirogacestat	• Estimate of 2-year overall survival
• To determine the effect of nirogacestat on ovarian cancer symptoms as measured by FOSI	• Change from baseline in FOSI
• To determine the duration of response to nirogacestat in participants who achieve CR or PR	• Duration of response, defined as time from response (CR + PR by RECIST v1.1 criteria) to disease progression and/or death
• To describe the PK profile of nirogacestat	• C_{max}, C_{trough}, and other PK parameters as data allow
Safety Objectives	**Safety Endpoints**
• To characterize the safety and tolerability of nirogacestat at a dose of 150 mg PO BID in adult patients	• Incidence of TEAEs • Changes in clinical laboratory parameters, vital signs, physical examination findings, and ECGs • Toxicities graded by NCI CTCAE v5.0
Exploratory Objectives	**Exploratory Endpoints**
• To detect *FOXL2* C134W mutation as well as other genomic alterations and correlate these with response	• NGS status in baseline tumor tissue
• To detect NICD and candidate biomarkers of response, and to correlate nirogacestat exposure with response	• Change from baseline in blood levels of hormones and tumor markers • inhibin A, inhibin B, FSH, estradiol, CA-125, MIS/AMH • ctDNA • Baseline NICD expression in tumor tissue

AMH, anti-Müllerian hormone; CA-125, cancer antigen 125; CR, complete response; CTCAE, Common Terminology Criteria for Adverse Events; ctDNA, circulating tumor DNA; ECG, electrocardiogram; FOSI, Functional Assessment of Cancer Therapy – Ovarian Symptom Index; FOXL2, forkhead box transcription factor 2; FSH, follicle-stimulating hormone; MIS, Müllerian-inhibiting substance; NCI, National Cancer Institute; NGS, next generation sequencing; NICD, Notch intracellular domain; ORR, objective response rate; OvGCT, ovarian granulosa cell tumor; PFS-6, 6-month progression-free survival; PK, pharmacokinetic; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; TEAE, treatment-emergent adverse event.

SUMMARY

- While surgery is indicated in the initial treatment of OvGCT, these tumors have a tendency for late recurrence following surgery and platinum-based chemotherapy
- Currently there are no FDA-approved therapies for OvGCT, and in relapsed patients, chemotherapeutic regimens have shown only modest activity
- Notch signaling – which is dependent on the proteolytic activity of γ-secretase – appears to play an integral role in the molecular pathogenesis of OvGCT, suggesting a mechanistic rationale for targeting γ-secretase inhibition in OvGCT
- This Phase 2, open-label, multicenter study will evaluate efficacy, safety, tolerability, and pharmacokinetics of oral nirogacestat, a potent inhibitor of γ-secretase, in the treatment of OvGCT

REFERENCES

1. Schumer ST, Cannistra SA. *J Clin Oncol*. 2003;21:1180–9.
2. Brown et al. *Cancer*. 2014;120:344-51.
3. Ray-Coquard et al. *JAMA Oncol*. 2020;6:1923-1930.
4. van Meurs et al. *Acta Obstet Gynecol Scand*. 2015;94:1269-75.
5. Li et al. *J Ovarian Res*. 2018;11:13.
6. Kopan R, Ilagan MXG. *Cell*. 2009;137:216–233.
7. McCaw et al. *Oncologist*. 2021;26:e608-e621.
8. Shang et al. *Cancer*. 2015;121:4088-96.

ACKNOWLEDGMENTS

This study is supported by SpringWorks Therapeutics, Inc. (Stamford, CT). Medical writing and editorial support were provided by Prescott Medical Communications Group (Chicago, IL) with financial support from SpringWorks Therapeutics.

PRESENTING AUTHOR DISCLOSURE

Panagiotis A. Konstantinopoulos, MD, has served as a consultant to AstraZeneca, Bayer, GSK, Alkermes, Kadmon, Bristol-Myers Squibb, IMV, Repare, Artios, Mersana, Immunogen, and Cardiff, and has received institutional support as a principal investigator from AstraZeneca, Bayer, Merck, Pfizer, GSK, Merck KGaA, and Eli Lilly.

Presented at the Society of Gynecologic Oncology 2023 Annual Meeting on Women's Cancer, March 26, 2023, Tampa, FL.