Efficacy of Nirogacestat in Patients With Poor Prognostic Factors for Desmoid Tumors: Analyses From the Randomized Phase 3 DeFi Study

ABSTRACT NUMBER: 11556

Bruno Vincenzi, MD, PhD[1]; Nam Quoc Bui, MD[2]; Palma Dileo, MD[3]; Noah Federman, MD[4]; Richard F. Riedel, MD[5]; Brian A. Van Tine, MD, PhD[6]; Vincent Amoruccio, PhD[7]; Allison Lim, PharmD[7]; Ana B. Oton, MD[7]; Bernd Kasper, MD, PhD[8]

[1]Policlinico Universitario Campus Bio-Medico, Rome, Italy; [2]Stanford University, Stanford, CA, USA; [3]University College London Hospitals NHS Foundation Trust, London, United Kingdom; [4]UCLA David Geffen School of Medicine, Los Angeles, CA, USA; [5]Duke Cancer Institute, Duke University, Durham, NC, USA; [6]Washington University, Saint Louis, MO, USA; [7]SpringWorks Therapeutics, Inc., Stamford, CT, USA; [8]University of Heidelberg, Mannheim University Medical Center, Mannheim, Germany

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

INTRODUCTION

- Desmoid tumors (DT) are rare, locally aggressive, soft-tissue tumors with a highly unpredictable natural course that exert substantial symptom burden on patients, including pain and functional limitation[1,2]
- Nirogacestat, a targeted gamma secretase inhibitor, is the only FDA-approved treatment for adults with progressing DT who require systemic treatment[3,4]
- In the phase 3 DeFi study,[5] nirogacestat demonstrated significant and clinically meaningful improvement versus placebo in the primary and key secondary endpoints of:
 - Progression-free survival (PFS; HR, 0.29 [95% CI, 0.15–0.55]; *P*<0.001)
 - Objective response rate (ORR; 41% versus 8%; *P*<0.001)
 - Patient-reported outcomes (pain, DT-specific symptom burden, physical and role functioning, and overall quality of life; all *P*≤0.01)
- Historically, prognosis of DT has potentially been dependent on multiple patient- and tumor-related factors (**Figure 1**)[6-13]
 - Larger tumor size, younger age, *CTNNB1* gene mutation, and pain at baseline have been associated with a poor prognosis in DT following active surveillance and treatments including surgical resection, radiotherapy, and systemic therapy not indicated specifically for DT[6-13]

Figure 1. Poor prognosis factors in patients with desmoid tumors

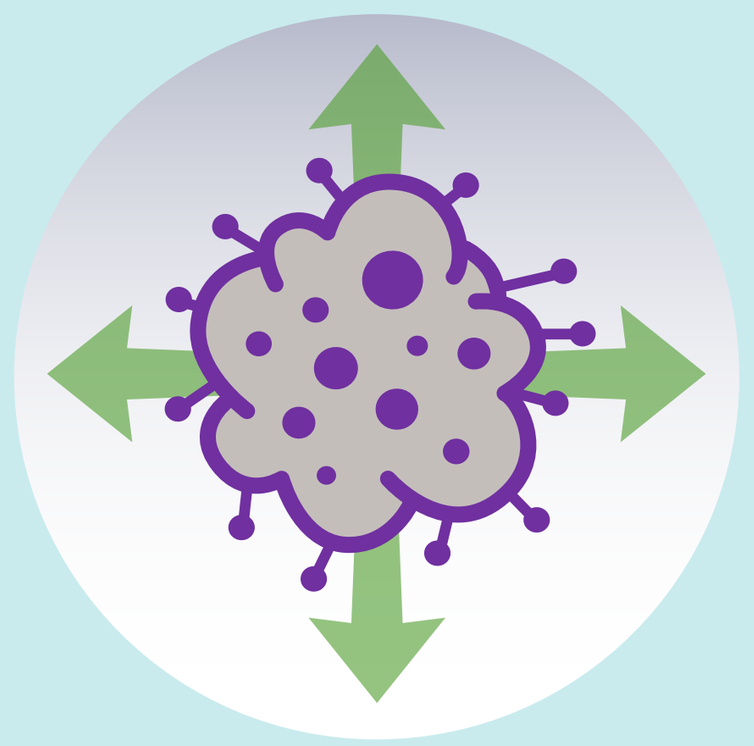

Larger Tumor Size

Larger size of primary tumors (>10 cm) has been associated with lower rates of 5-year recurrence-free survival and poorer local control[6,13]

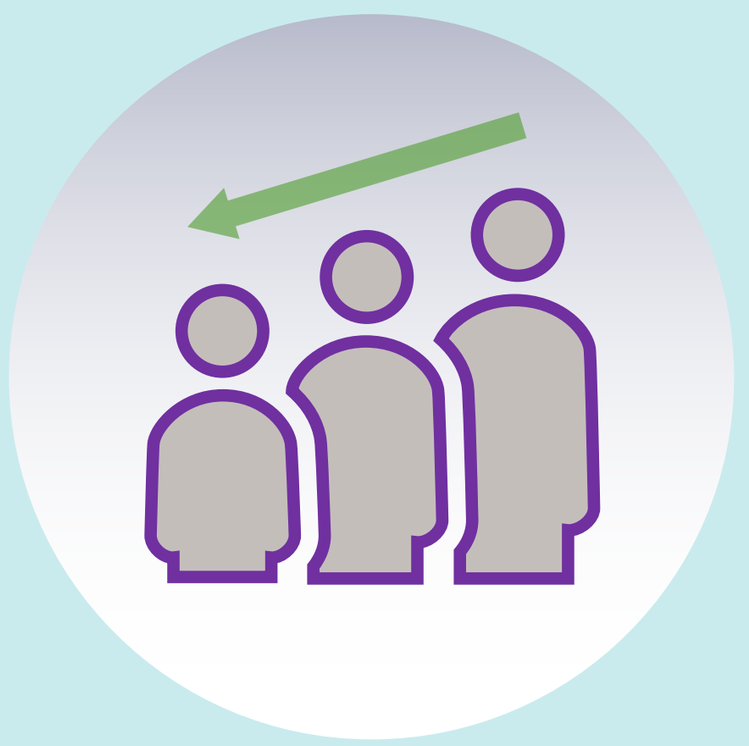

Younger Patient Age

Although the association between age and DT recurrence has not been fully corroborated,[14] some evidence has suggested younger patients (eg, ≤30 years) could potentially have a higher risk of recurrence and worse local control than older patients[6,7,13,15]

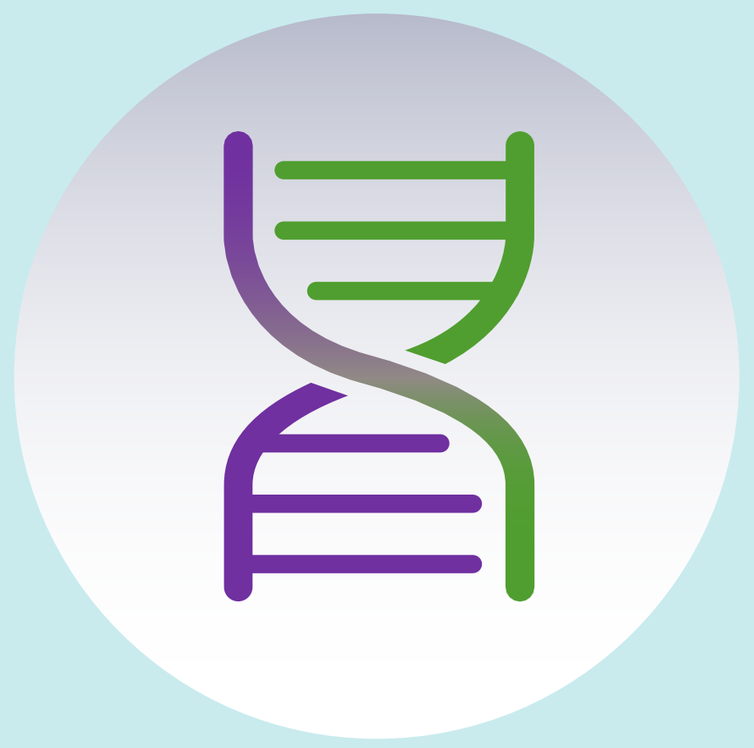

***CTNNB1* Mutation**

- Studies have shown an association between any *CTNNB1* mutation and inferior 5-year recurrence-free survival[9]
- Of patients with *CTNNB1* gene mutations, T41A and S45F mutations are the most common[10]
 - S45F mutation may be associated with poorer outcomes compared with T41A mutation[8,10,16]

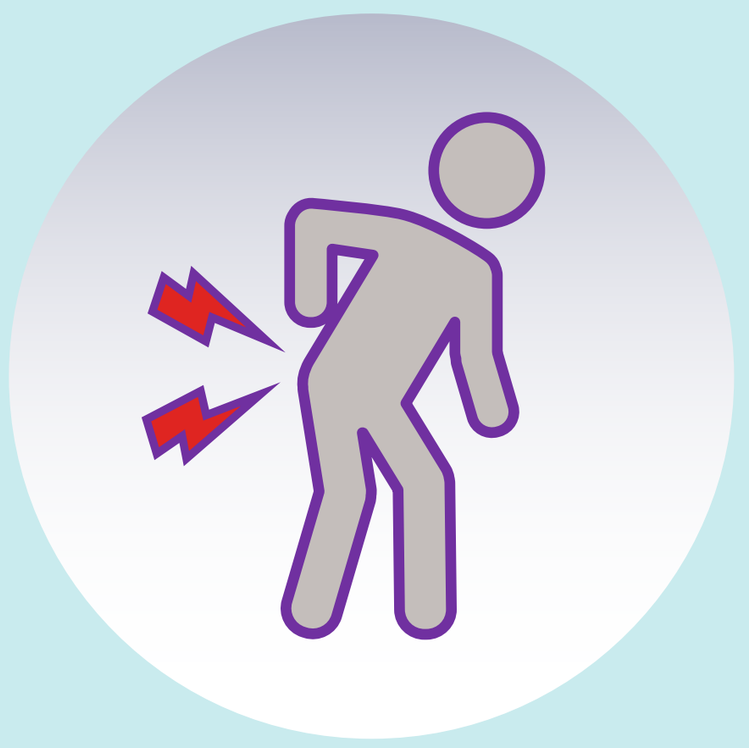

Presence of Pain

Patients reporting pain at baseline have been shown to have higher disease progression rates and lower event-free survival compared with those without pain[11,12]

OBJECTIVE

- A post hoc analysis of the DeFi study was conducted to assess the effect of nirogacestat in subgroups of patients with DT who have risk factors associated with poor prognosis

METHODS

- DeFi (NCT03785964) was a global, multicenter, double-blind, pivotal phase 3 study to determine the efficacy, safety, and tolerability of nirogacestat in adults with progressing DT
- Patients were randomized 1:1 to nirogacestat 150 mg (n=70) or placebo (n=72), taken twice-daily in 28-day cycles
- Post hoc analyses of PFS and ORR were performed in subgroups of patients with demographic- and tumor-related characteristics at baseline associated with poor prognosis:
 - >10 cm tumor size[6,13]
 - ≤30 years patient age[6,7,13,15]
 - Presence of *CTNNB1* mutation (S45F, T41A)[9,10]
 - Presence of pain at baseline as determined by an average pain intensity (API) score of >0 on the Brief Pain Inventory–Short Form (BPI-SF)[11,12]

RESULTS

- PFS improvement favored nirogacestat compared with placebo in patients with larger tumor size, younger age, *CTNNB1* mutation, and presence of pain at baseline; nirogacestat was favored over placebo in both *CTNNB1* mutation types (S45F or T41A) (**Figure 2**)
- ORR improvement favored nirogacestat versus placebo regardless of the poor prognostic factor (**Figure 3**)

Figure 2. Progression-free survival in poor prognostic subgroups

Factor	Hazard Ratio (95% CI)	Nirogacestat Events/Total n/N	Placebo Events/Total n/N
Baseline tumor size			
>10 cm	0.32 (0.13, 0.80)	6/29	21/42
Age			
≤30 y	0.21 (0.08, 0.60)	5/30	16/27
***CTNNB1* mutation**			
S45F	0.18 (0.02, 1.46)	1/13	8/18
T41A	0.39 (0.14, 1.11)	5/24	12/22
Baseline pain[a]			
BPI-SF API >0	0.21 (0.09, 0.52)	6/47	23/46

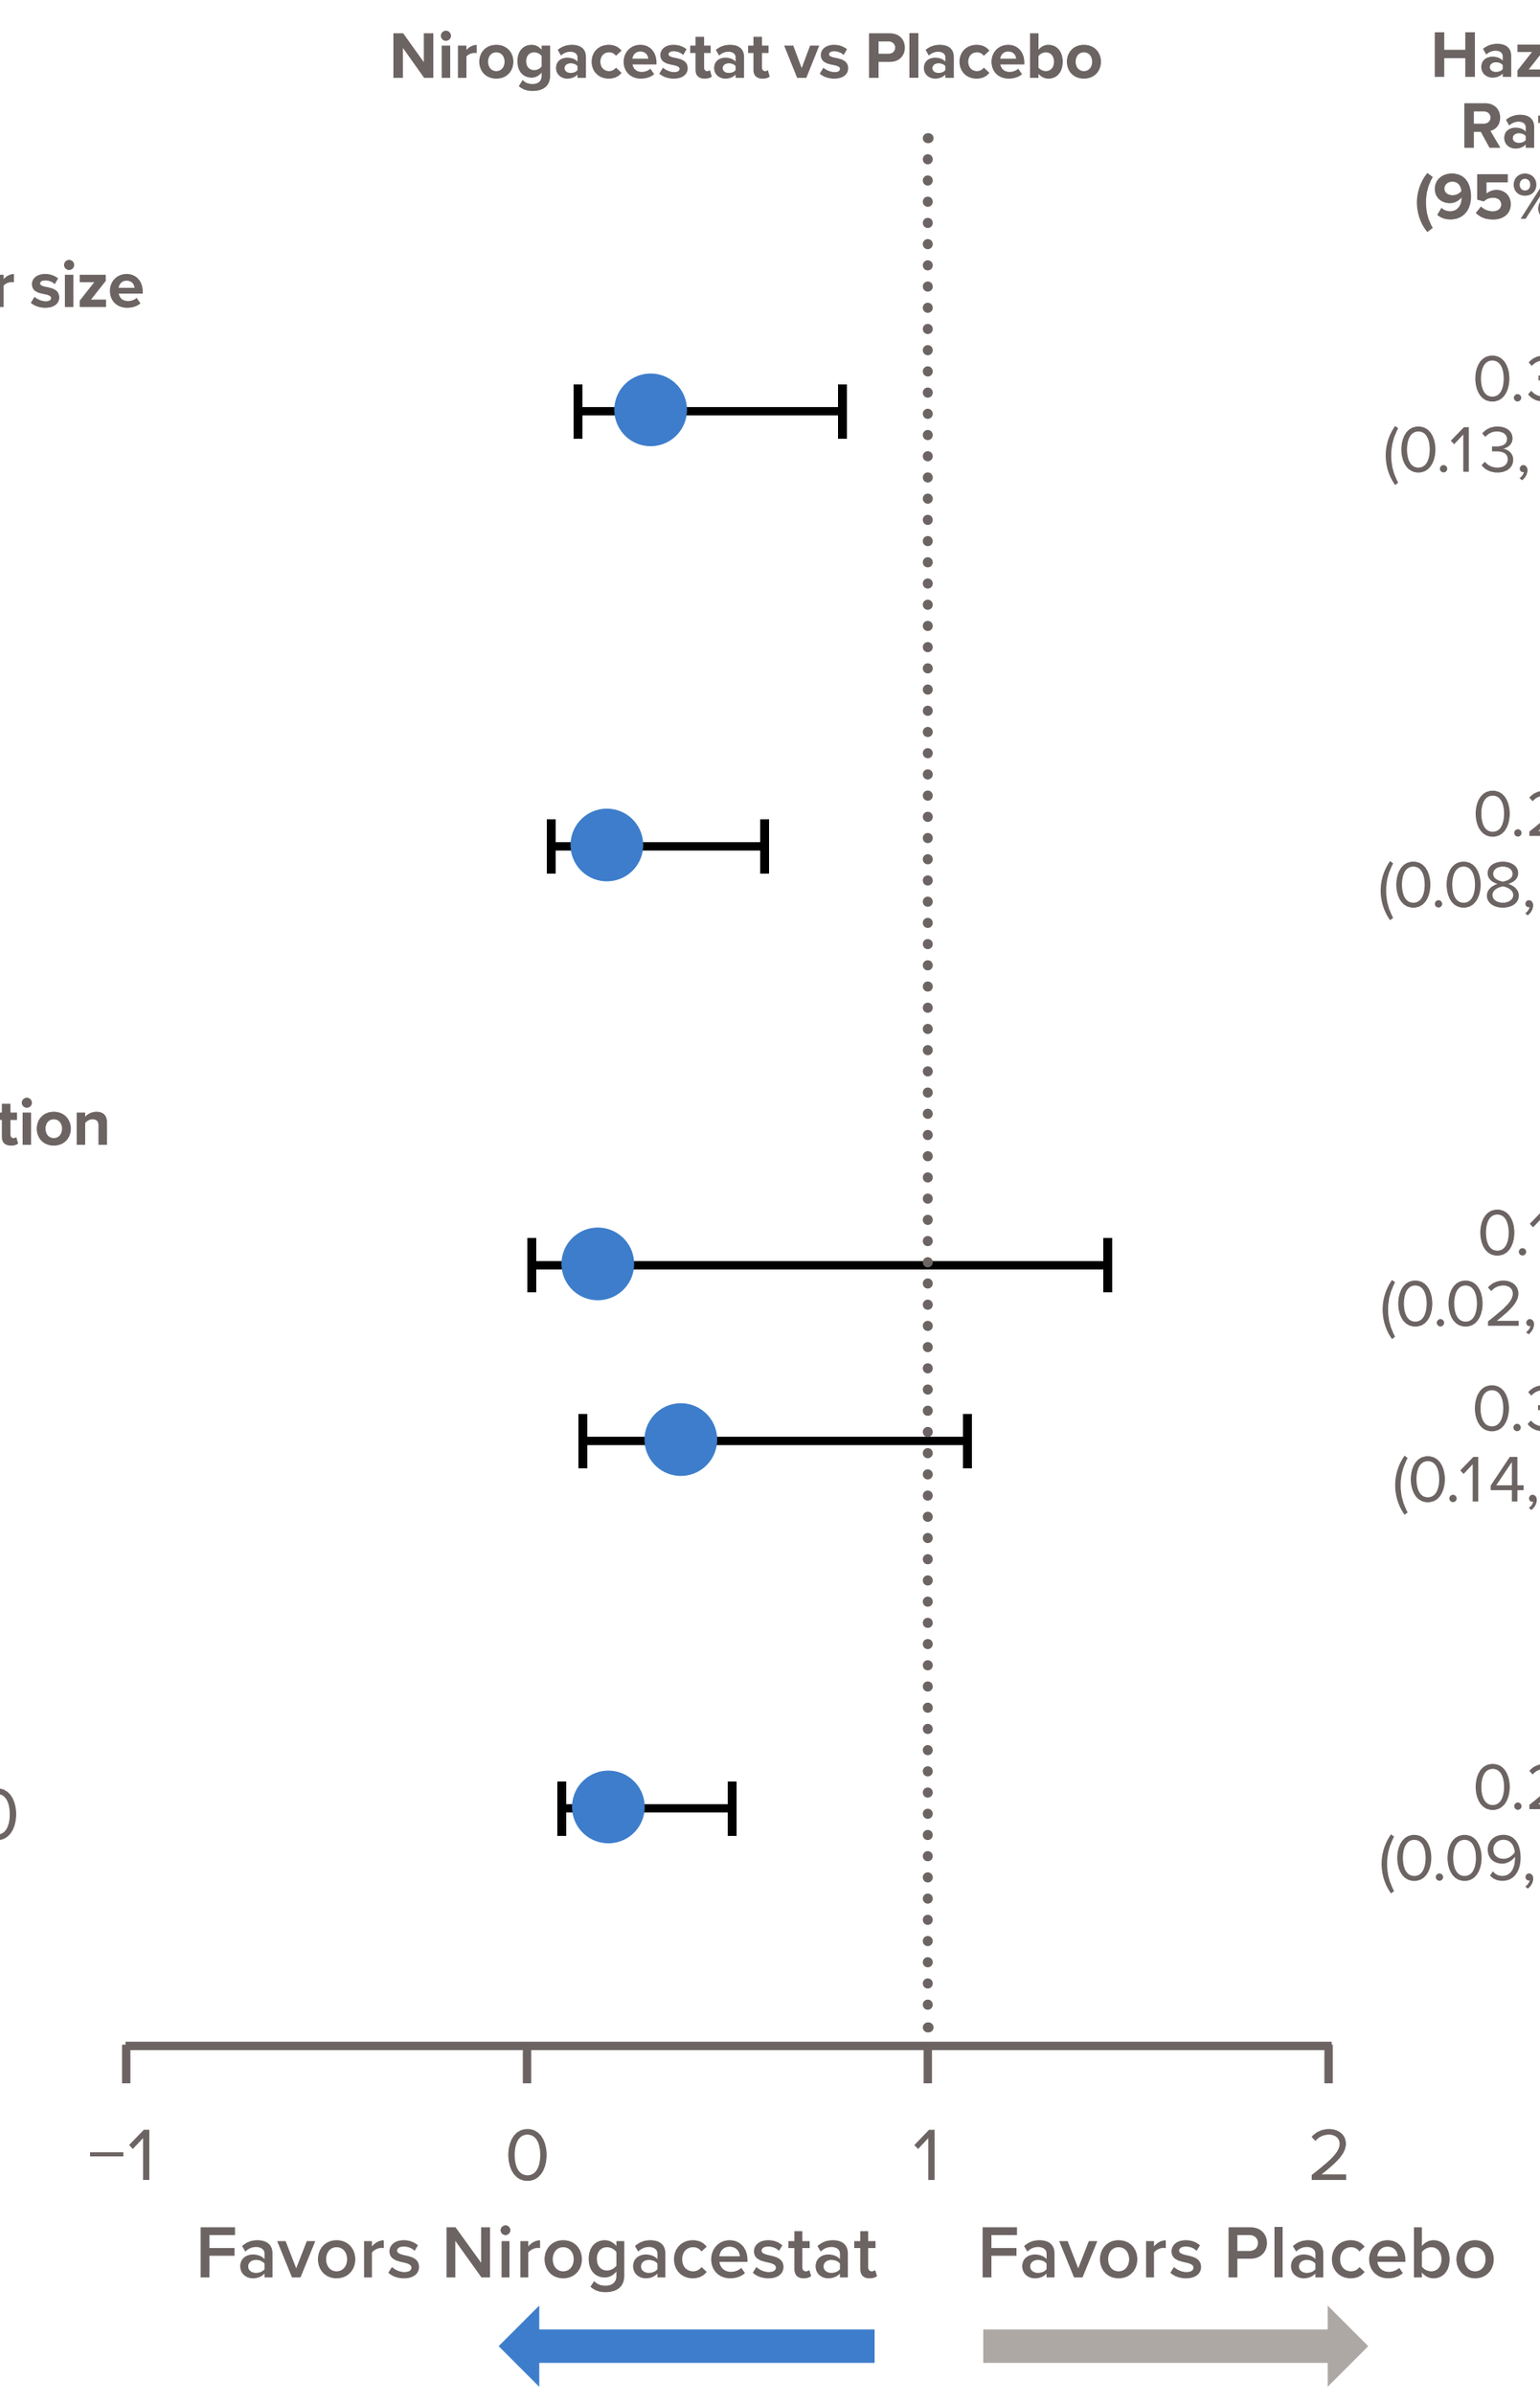
Nirogacestat vs Placebo
−1
0
1
2
Favors Nirogacestat
Favors Placebo

[a]Pain was assessed using BPI-SF item 3 (worst pain in the past 24 hours). API was calculated using daily scores from up to a 7-day period prior to each visit; scores ranged from 0–10, with higher scores indicating worse pain.
For PFS, hazard ratio was estimated from the stratified Cox proportion hazards model using the exact method for ties, stratified by tumor location. PFS was calculated as the earliest date of death, progression, or censoring from randomization. Patients who were discontinued early by investigators for clinical progression but could not be verified as events, were censored. Patients who did not progress or die by the date of the last valid CT/MRI assessment were censored.
API, average pain intensity; BPI-SF, Brief Pain Inventory–Short Form; CI, confidence interval; CT, computed tomography; MRI, magnetic resonance imaging; PFS, progression-free survival.

Figure 3. Objective response rate in poor prognostic subgroups

Factor	Risk Difference, %[a] (95% CI)	Nirogacestat n (%)	Placebo n (%)
Baseline tumor size			
>10 cm	18.1 (−0.5, 36.6)	8 (28)	4 (10)
Age			
≤30 y	36.3 (17.4, 55.2)	12 (40)	1 (4)
***CTNNB1* mutation**			
S45F	56.0 (27.5, 84.5)	8 (62)	1 (6)
T41A	24.2 (1.9, 46.6)	8 (33)	2 (9)
Baseline pain[b]			
BPI-SF API >0	33.9 (17.5, 50.2)	20 (43)	4 (9)

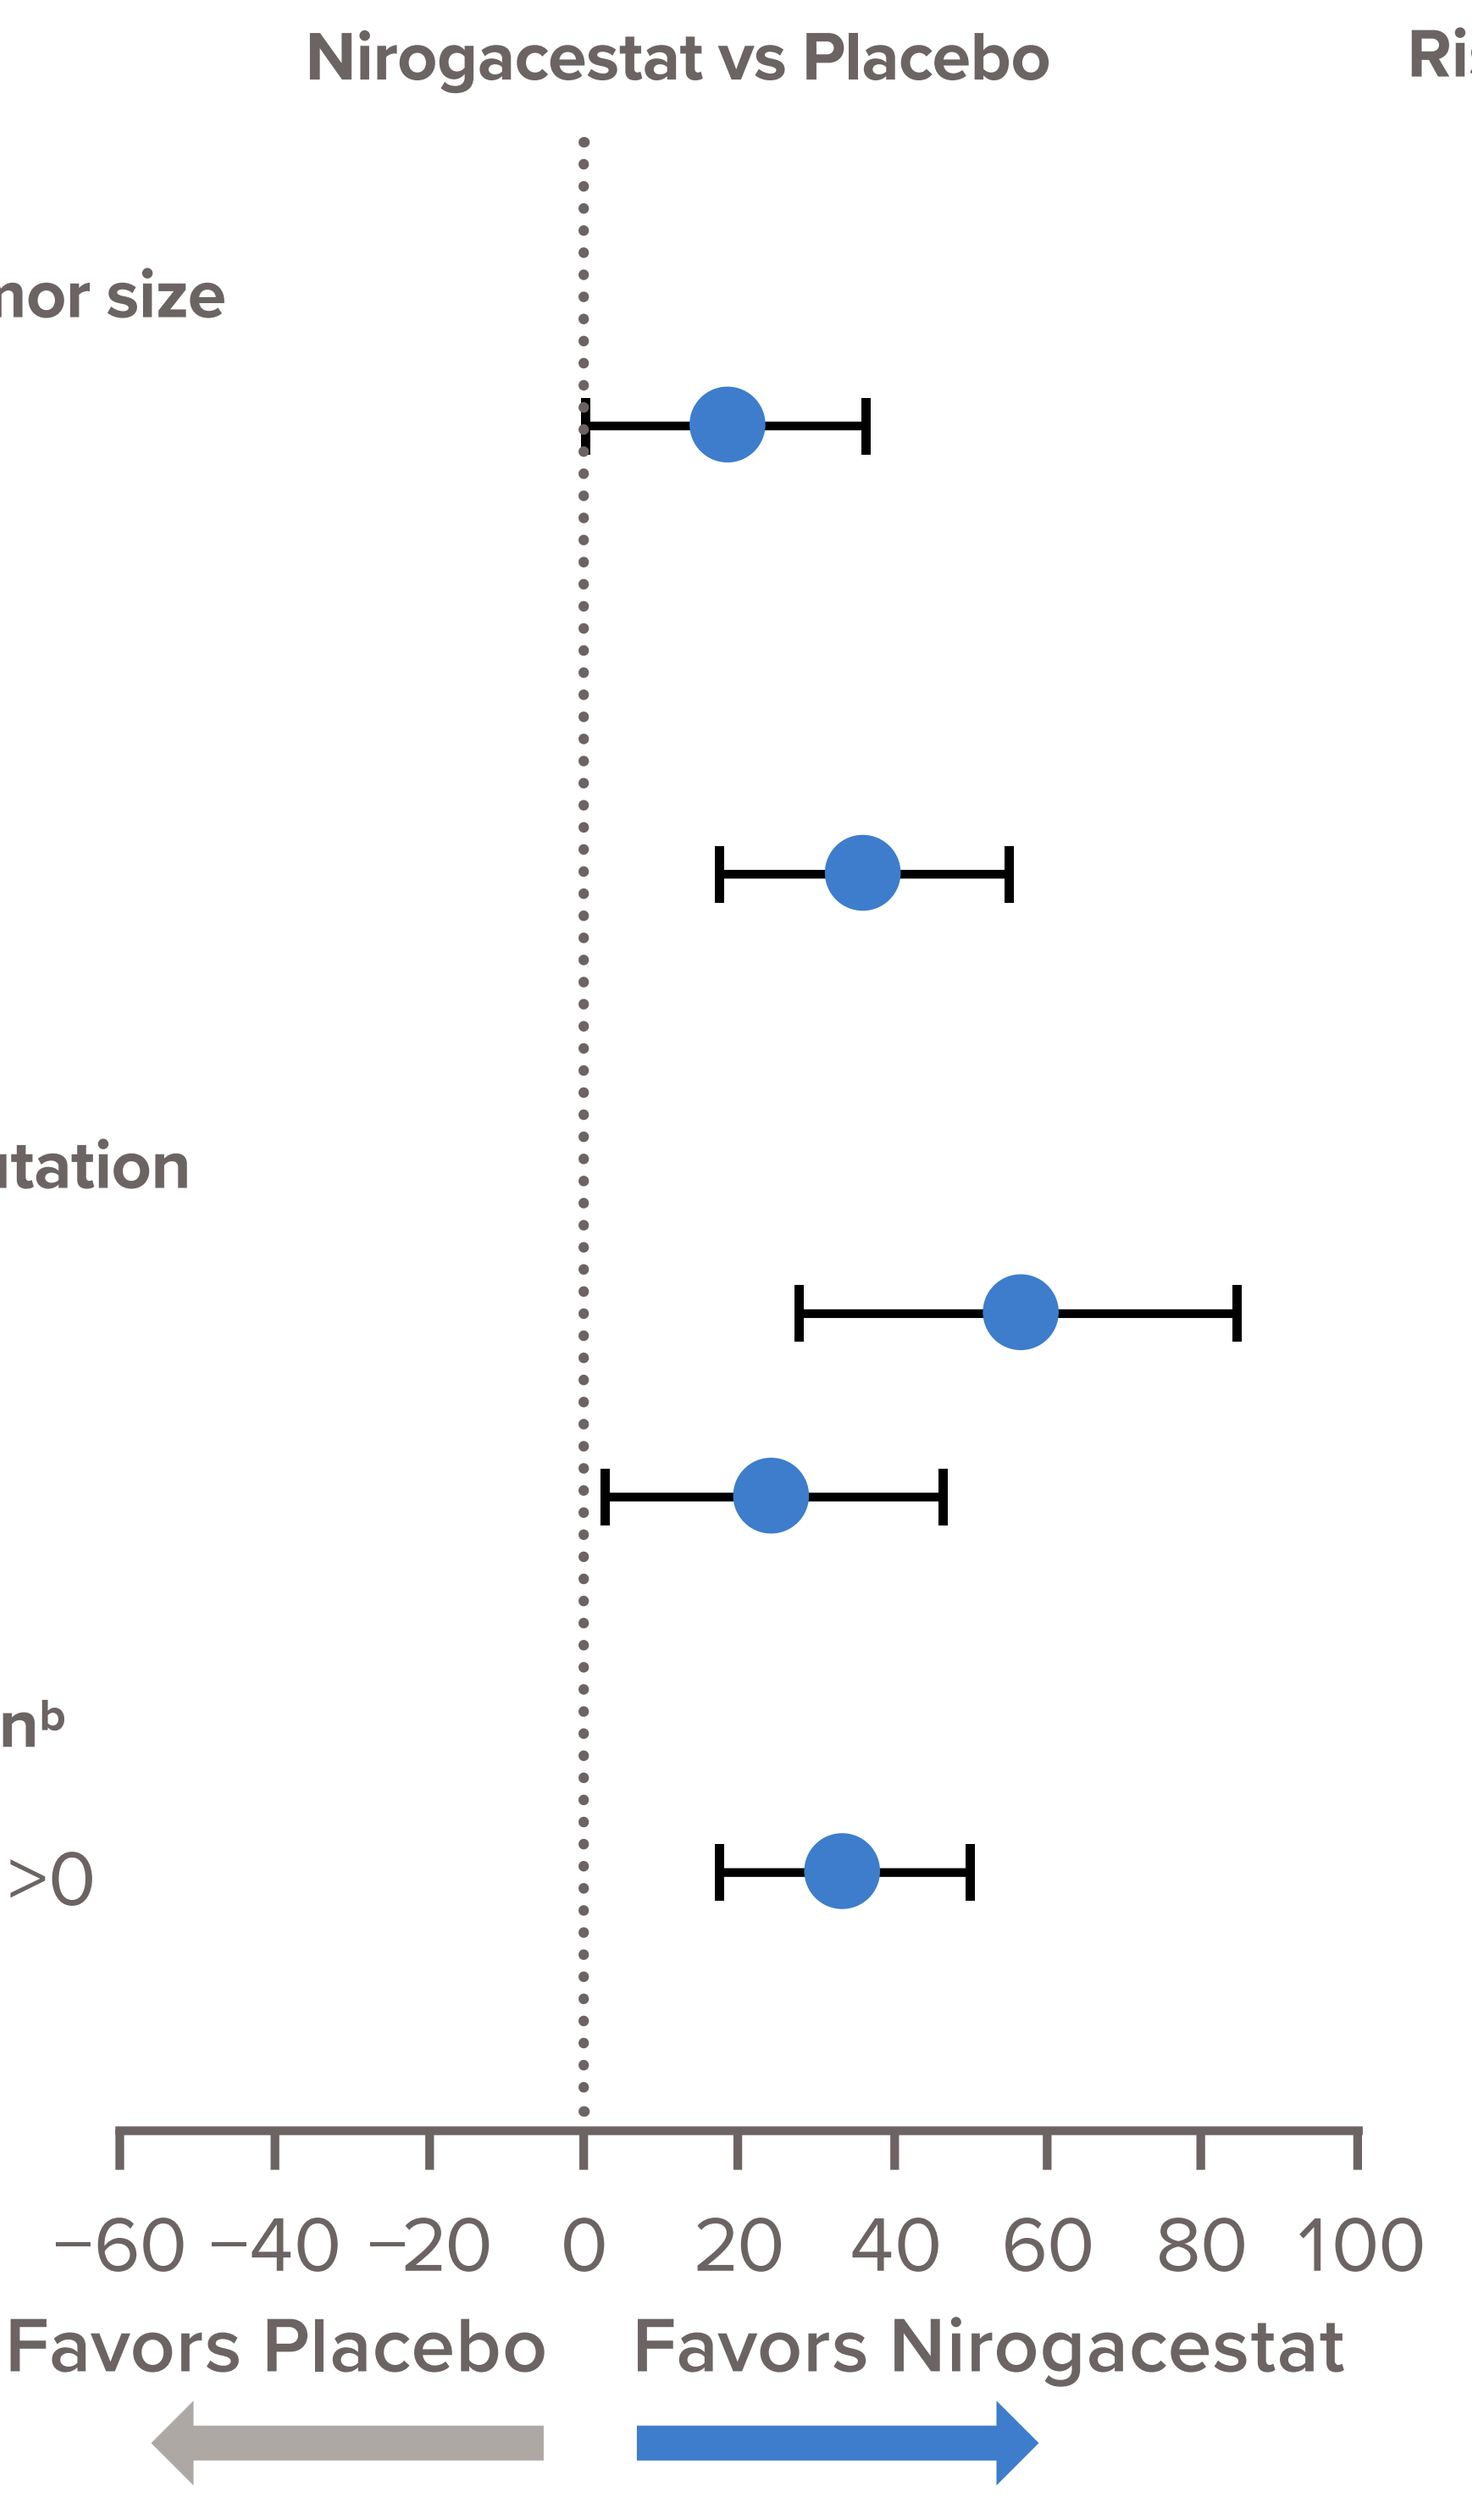
Nirogacestat vs Placebo
−60 −40 −20 0 20 40 60 80 100
Favors Placebo
Favors Nirogacestat

[a]The difference between the ORR in nirogacestat and placebo; a risk difference >0 favors nirogacestat.
[b]Pain was assessed using BPI-SF item 3 (worst pain in the past 24 hours). API was calculated using daily scores from up to a 7-day period prior to each visit; scores ranged from 0–10, with higher scores indicating worse pain.
API, average pain intensity; BPI-SF, Brief Pain Inventory–Short Form; CI, confidence interval; ORR, objective response rate.

CONCLUSIONS

- Nirogacestat demonstrated consistent improvement in PFS and ORR versus placebo in patients with characteristics associated with poor prognosis in DT, including larger tumor size, younger age, *CTNNB1* gene mutation, and presence of pain at baseline
 - Results were consistent with the overall DeFi population[5]
- These results indicate that nirogacestat can provide substantial benefit in patients with characteristics that have been historically associated with poor prognosis following active surveillance and treatments including surgical resection, radiotherapy, and systemic therapy

REFERENCES

1. Bektas M, et al. *Adv Ther.* 2023;40(9):3697-3722.
2. Kasper B, et al. *Ann Oncol.* 2017;28(10):2399-2408.
3. OGSIVEO (nirogacestat) [package insert]. Stamford, CT: SpringWorks Therapeutics, Inc: 2023. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/217677s000lbl.pdf
4. US Food and Drug Administration website. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-nirogacestat-desmoid-tumors. Revised November 28, 2023. Accessed April 25, 2024.
5. Gounder M, et al. *N Engl J Med.* 2023;388(10):898-912.
6. Crago AM, et al. *Ann Surg.* 2013;258(2):347-53.
7. Lev D, et al. *J Clin Oncol.* 2007;25(13):1785-1791.
8. Colombo C, et al. *Cancer.* 2013;119(20):3696-3702.
9. Dômont J, et al. *Br J Cancer.* 2010;102(6):1032-1036.
10. Timbergen MJM, et al. *Ann Surg.* 2021;273(6):1094-1101.
11. Cuomo P, et al. *BMC Cancer.* 2021;21(1):437.
12. Penel N, et al. *Int J Cancer.* 2023;153(2):407-416.
13. Bishop AJ, et al. *Cancer.* 2020;126(14):3265-3273.
14. Pignatti G, et al. *Clin Orthop Relat Res.* 2000;375:207-213.
15. Sørensen A, et al. *Acta Orthop Scand.* 2002;73(2):213-219.
16. Lazar AJF, et al. *Am J Pathol.* 2008;173(5):1518-1527.

ACKNOWLEDGMENTS

This presentation was supported by SpringWorks Therapeutics, Inc. Medical writing support was provided by Saba Choudhary, PhD, from Prescott Medical Communications Group, a Citrus Health Group, Inc. company (Chicago, IL), and was funded by SpringWorks Therapeutics, Inc.

Presented at the Annual Meeting of the American Society of Clinical Oncology • May 31 – June 4, 2024; Chicago, IL